USE OF AN EFFICIENT LC AND SPE METHOD DEVELOPMENT APPROACH IN THE ANALYSIS OF TERFENADINE, FEXOFENADINE, AND AZACYCLONOL

Erin Chambers, Jessalynn Wheaton, and Diane M. Diehl
Waters Corporation , Milford, Massachusetts 01757 USA

BACKGROUND

Utilizing ACQUITY UPLC® Technology with triple quadrupole MS detection enhances the selectivity, sensitivity, and through-put in quantitative bioanalytical studies. Detection limits for these methods are being driven lower and lower as drugs become more potent. For this reason, methods with highest sensitivity possible must be developed. Additionally, this must happen under increasingly tight deadlines in order to shorten the drug development cycle. We have developed a straightforward, streamlined approach to efficient LC method development. The use of this approach is demonstrated in the generation of a sensitive and selective method for terfenadine and two of it's metabolites, fexofenadine and azacyclonol. Fexofenadine, in particular, has gained importance as the commonly prescribed anti-allergic Allegra®. The structures for all analytes are shown in Figure 1.

ANALYTICAL CHALLENGES

Metabolites and their parent compounds can differ dramatically in chemical properties, making it difficult to develop a chromatographic method which is optimal for all analytes. For example, a non-polar basic parent drug can easily undergo biotransformation into basic metabolites as well as polar acidic metabolites. The LC method must then provide adequate sensitivity for low levels of all these types of compounds, as well as resolution of structurally similar metabolites from the parent drug.

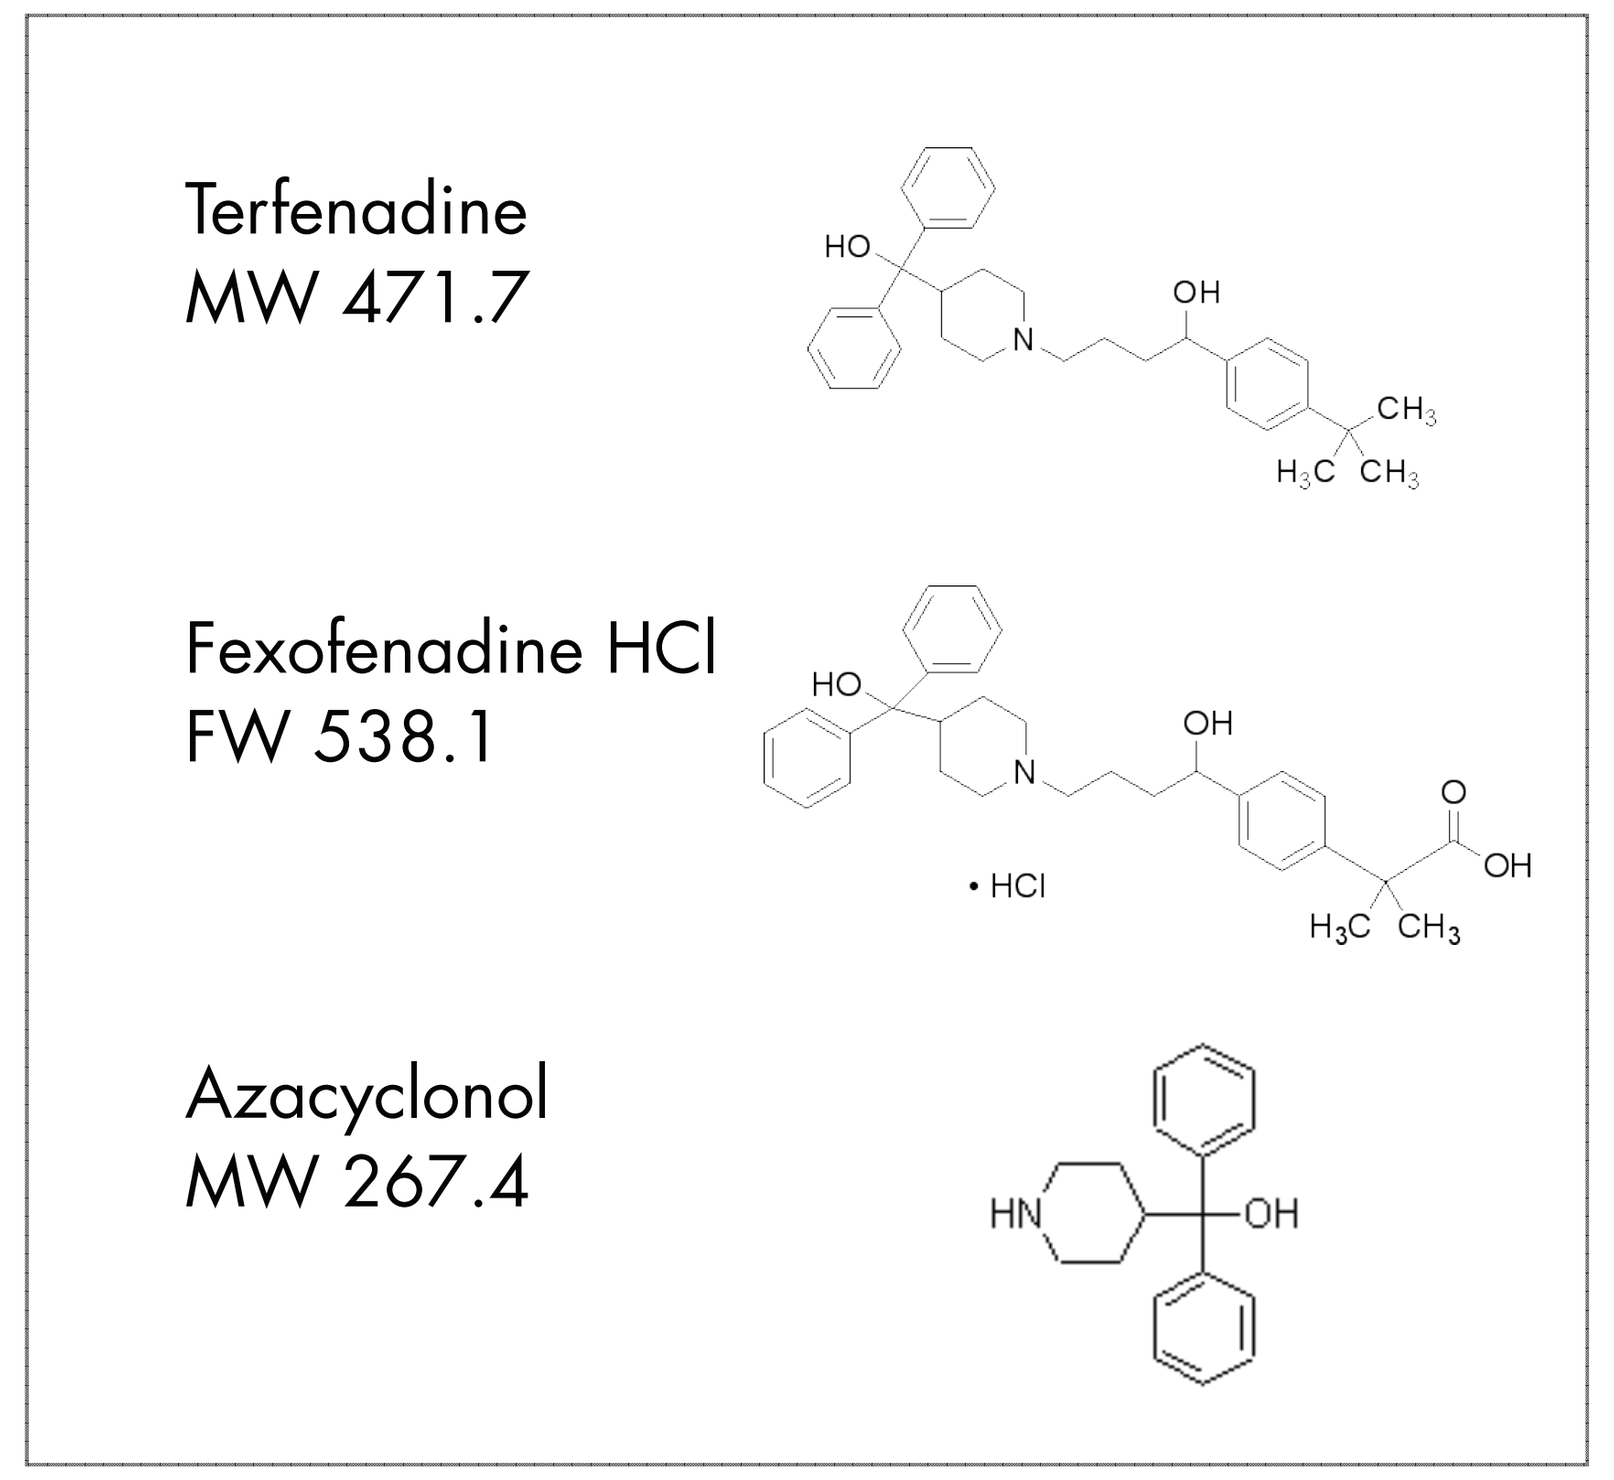

Terfenadine
MW 471.7
Fexofenadine HCl
FW 538.1
Azacyclonol
MW 267.4

Figure 1: Structures of terfenadine, fexofenadine, and azacyclonol.

EXPERIMENTAL

A screening approach was utilized to choose the best starting chromatographic conditions. Four different column chemistries were screened at high and low pH using two organic modifiers. The resulting chromatograms were evaluated for peak shape, retention, and selectivity. From these data, the column chemistry, pH, and mobile phase modifier were chosen. The chosen screening gradient was later optimized to decrease run time.

LC CONDITIONS FOR SCREENING

LC System: ACQUITY UPLC® and Quattro Premier

Columns: ACQUITY UPLC® BEH C_{18} 2.1 X 50 mm, 1.7 µm
ACQUITY UPLC® BEH Shield RP_{18} 2.1 X 50 mm, 1.7 µm
ACQUITY UPLC® BEH Phenyl 2.1 X 50 mm, 1.7 µm
ACQUITY UPLC® HSS T3 2.1 X 50 mm, 1.8 µm

Column Temp: 50°C

Flow Rate: 600 µL/min
Mobile Phase A1: 0.1% HCOOH in H2O (~pH 2.7)
Mobile Phase A2: 0.1% NH_4OH in H2O (~pH 11)
Mobile Phase B1: MeOH
Mobile Phase B2: ACN
Gradient: 5-95% B in 2 min., hold for 0.5 min; return to initial conditions

MS CONDITIONS

MS System: Waters Micromass Quattro Premier™

Ionization Mode: ESI Positive

Capillary Voltage: 3.2 kV

Cone Voltage: 17 V, 31 V, and 47 V for azacyclonol, terfenadine, and fexofenadine, respectively

Desolvation Temp: 350°C

Desolvation Gas: 650 L/Hr

Source Temp: 125°C

Collision Energy: 13 eV, 28 eV, and 31 eV for azacyclonol, terfenadine, and fexofenadine, respectively

MRM Channels: terfenadine m/z 472.6->436.6, fexofenadine m/z 502.4->466.3, azacyclonol m/z 268.2->250.2

SPE STRATEGY AND CONDITIONS

A simple, straightforward SPE screening strategy was employed to facilitate the choice of the starting SPE sorbent and protocol. This approach relies on 4 mixed-mode Oasis® sorbents and 2 simple protocols. The Oasis® µElution Sorbent Selection plate, which contains 3 rows of each of the 4 mixed-mode sorbents, was employed to make it easier to carry out the SPE screening. Figure 2 summarizes the methodology.

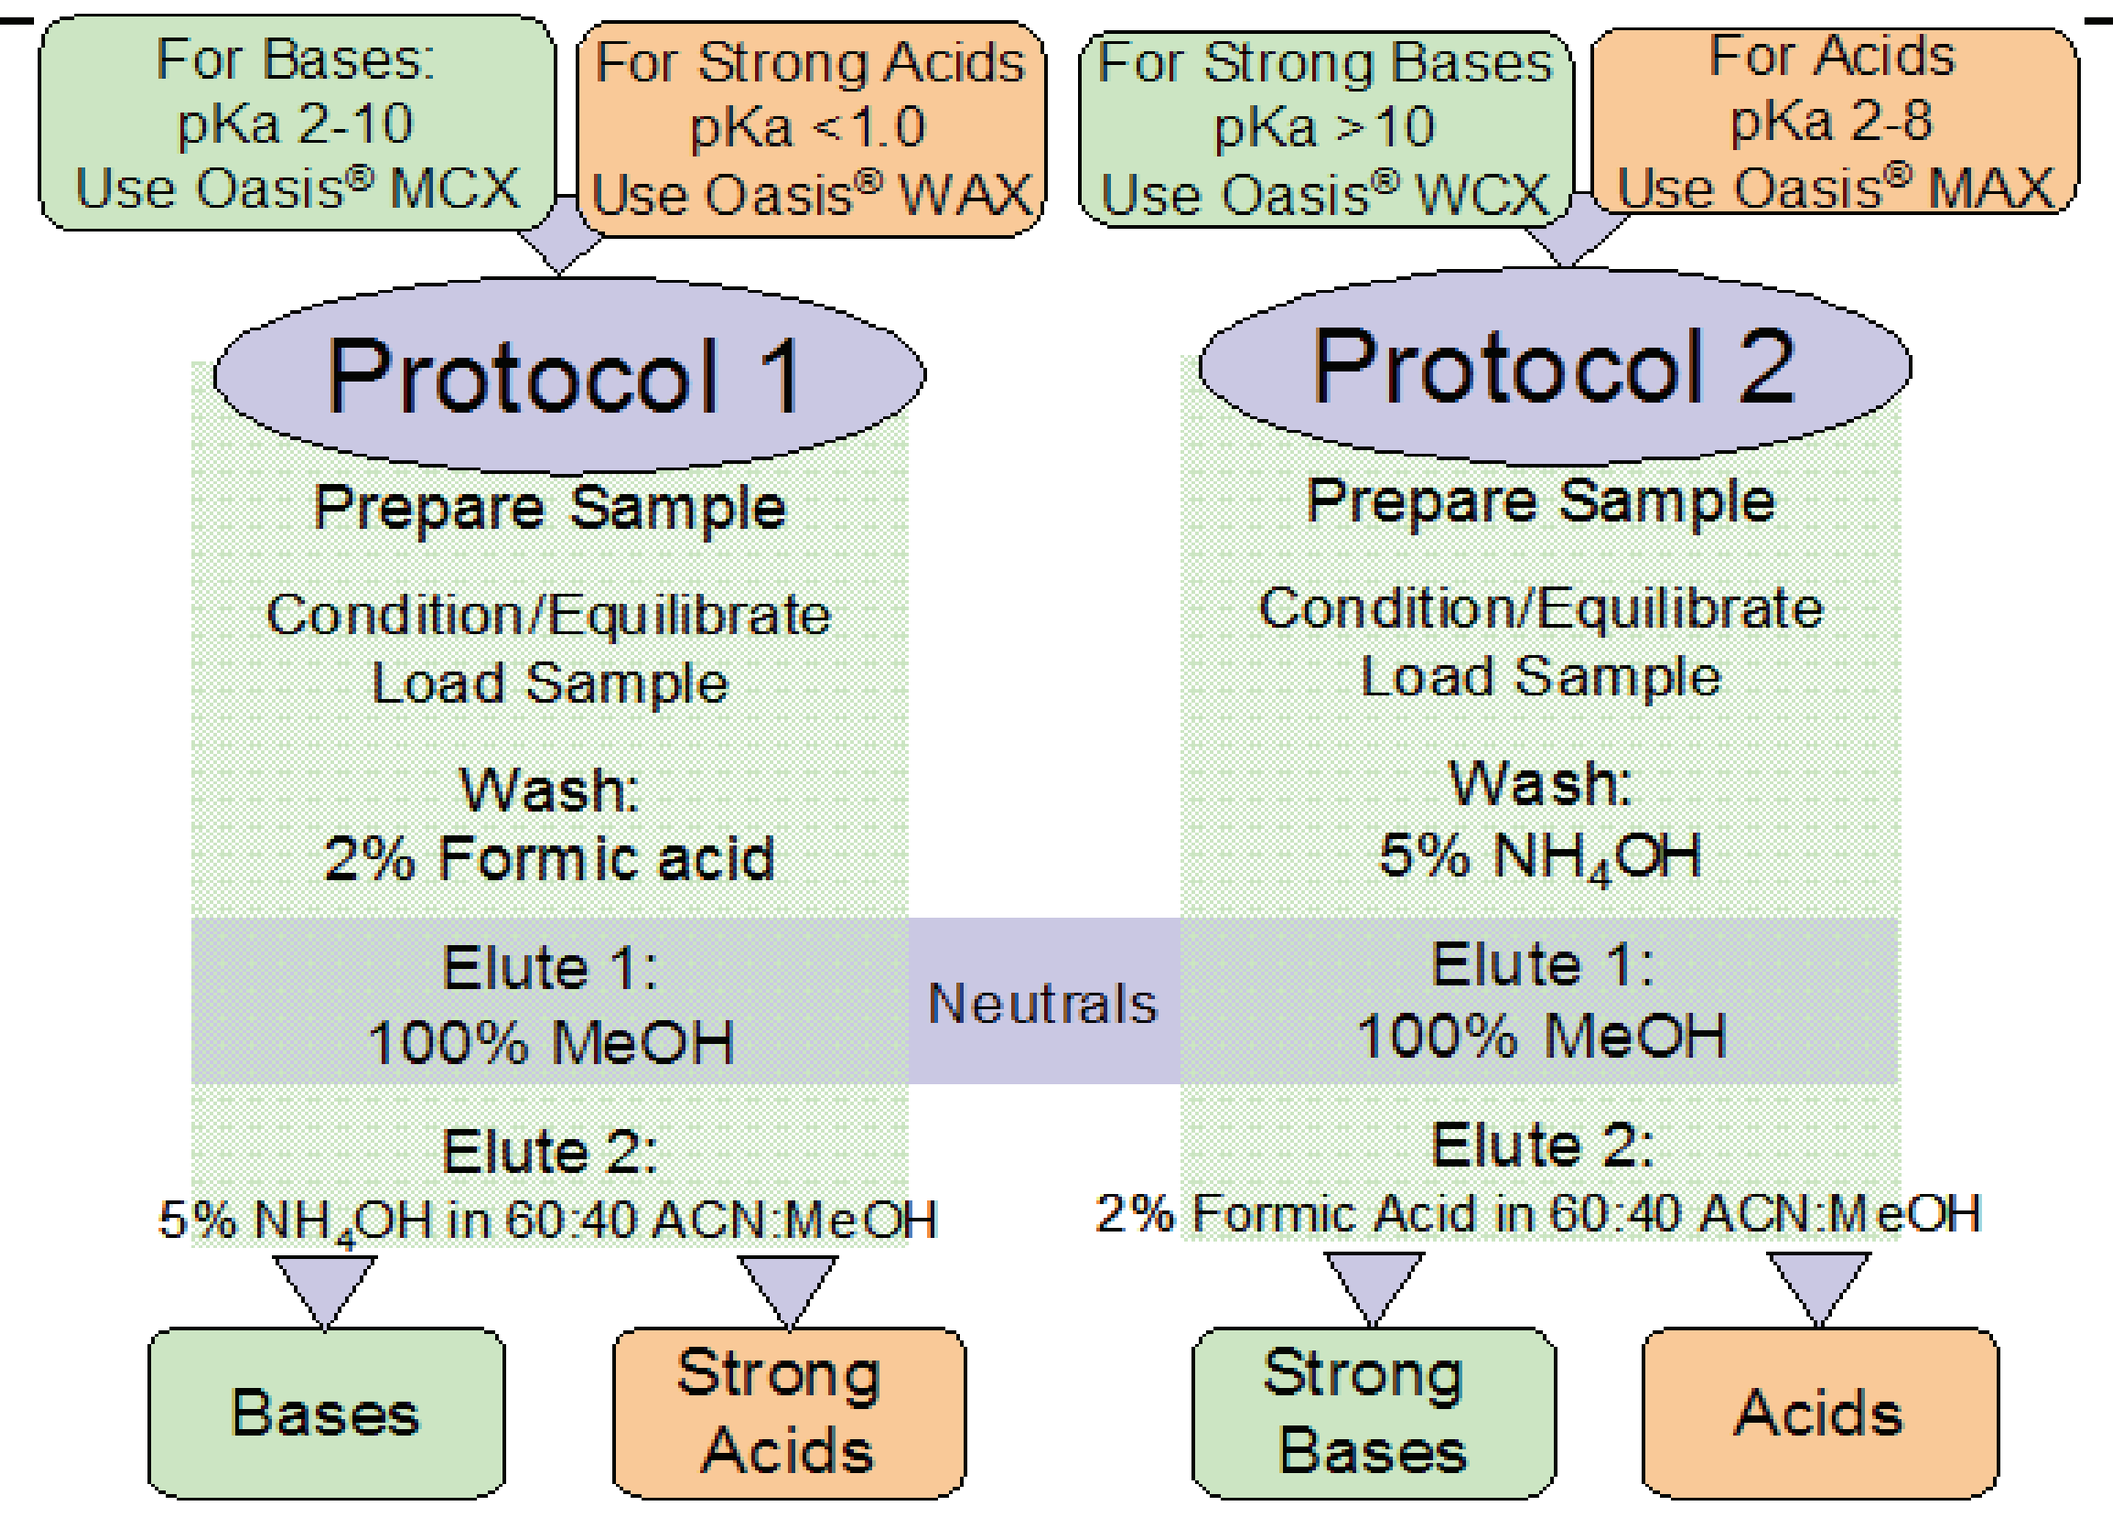

For Bases: pKa 2-10 Use Oasis® MCX
For Strong Acids pKa <1.0 Use Oasis® WAX
For Strong Bases pKa >10 Use Oasis® WCX
For Acids pKa 2-8 Use Oasis® MAX
Protocol 1
Prepare Sample
Condition/Equilibrate
Load Sample
Wash: 2% Formic acid
Elute 1: 100% MeOH
Elute 2: 5% NH4OH in 60:40 ACN:MeOH
Neutrals
Protocol 2
Prepare Sample
Condition/Equilibrate
Load Sample
Wash: 5% NH4OH
Elute 1: 100% MeOH
Elute 2: 2% Formic Acid in 60:40 ACN:MeOH
Bases
Strong Acids
Strong Bases
Acids

Figure 2. The Oasis® 2 X 4 Methodology: an SPE Screening Strategy

RESULTS AND DISCUSSION

CHROMATOGRAPHIC SCREENING

The chromatograms resulting from pH screening with ACN as the organic modifier are shown in Figure 3. Initial optimization was done to determine the MRM transitions and optimal mass spectrometry conditions for each compound. A TIC of the three transitions is shown in the figure. All columns were run at low pH (A). The C18, Shield RP18, and Phenyl columns were also run at high pH (B). (Note: the HSS T3 column contains a silica particle and cannot be run at high pH.) Chromatograms were evaluated with respect to peak shape, resolution of metabolites from each other and the parent drug, sensitivity, and selectivity. Selectivity shifts were observed for fexofenadine and azacyclonol at high versus low pH, as well as dramatic peak shape differences on the phenyl column for the azacyclonol metabolite. Retention of the basic, non-polar compound, terfenadine is dramatically affected by change in pH.

Notice that terfenadine is more strongly retained on the C_{18} column at high pH when it is in its neutral state. A significant increase in MS sensitivity (almost 3X) is also observed for this analyte under high pH conditions. Terfenadine now elutes in a higher percentage organic. Droplets which are higher in organic composition are more efficiently desolvated in the MS source, resulting in increased sensitivity. We chose low pH mobile phase due to excellent peak shape for all compounds and improved sensitivity for several compounds. We next screened all column chemistries using two organic modifiers. The chromatograms resulting from organic modifier screening are shown in Figure 4. All columns were run at low pH with MeOH (A) and ACN (B). All three compounds chromatograph as well-resolved, narrow peaks with both MeOH and ACN, with the exception of terfenadine on the phenyl column. However, sensitivity is slightly higher using MeOH as the organic modifier. The final step is choosing the optimal column chemistry. Examination of the low pH, MeOH chromatograms indicates that the best sensitivity for all compounds is achieved with the HSS T3 column. We optimized the chromatographic method using the HSS T3 column at low pH, with MeOH as the organic modifier.

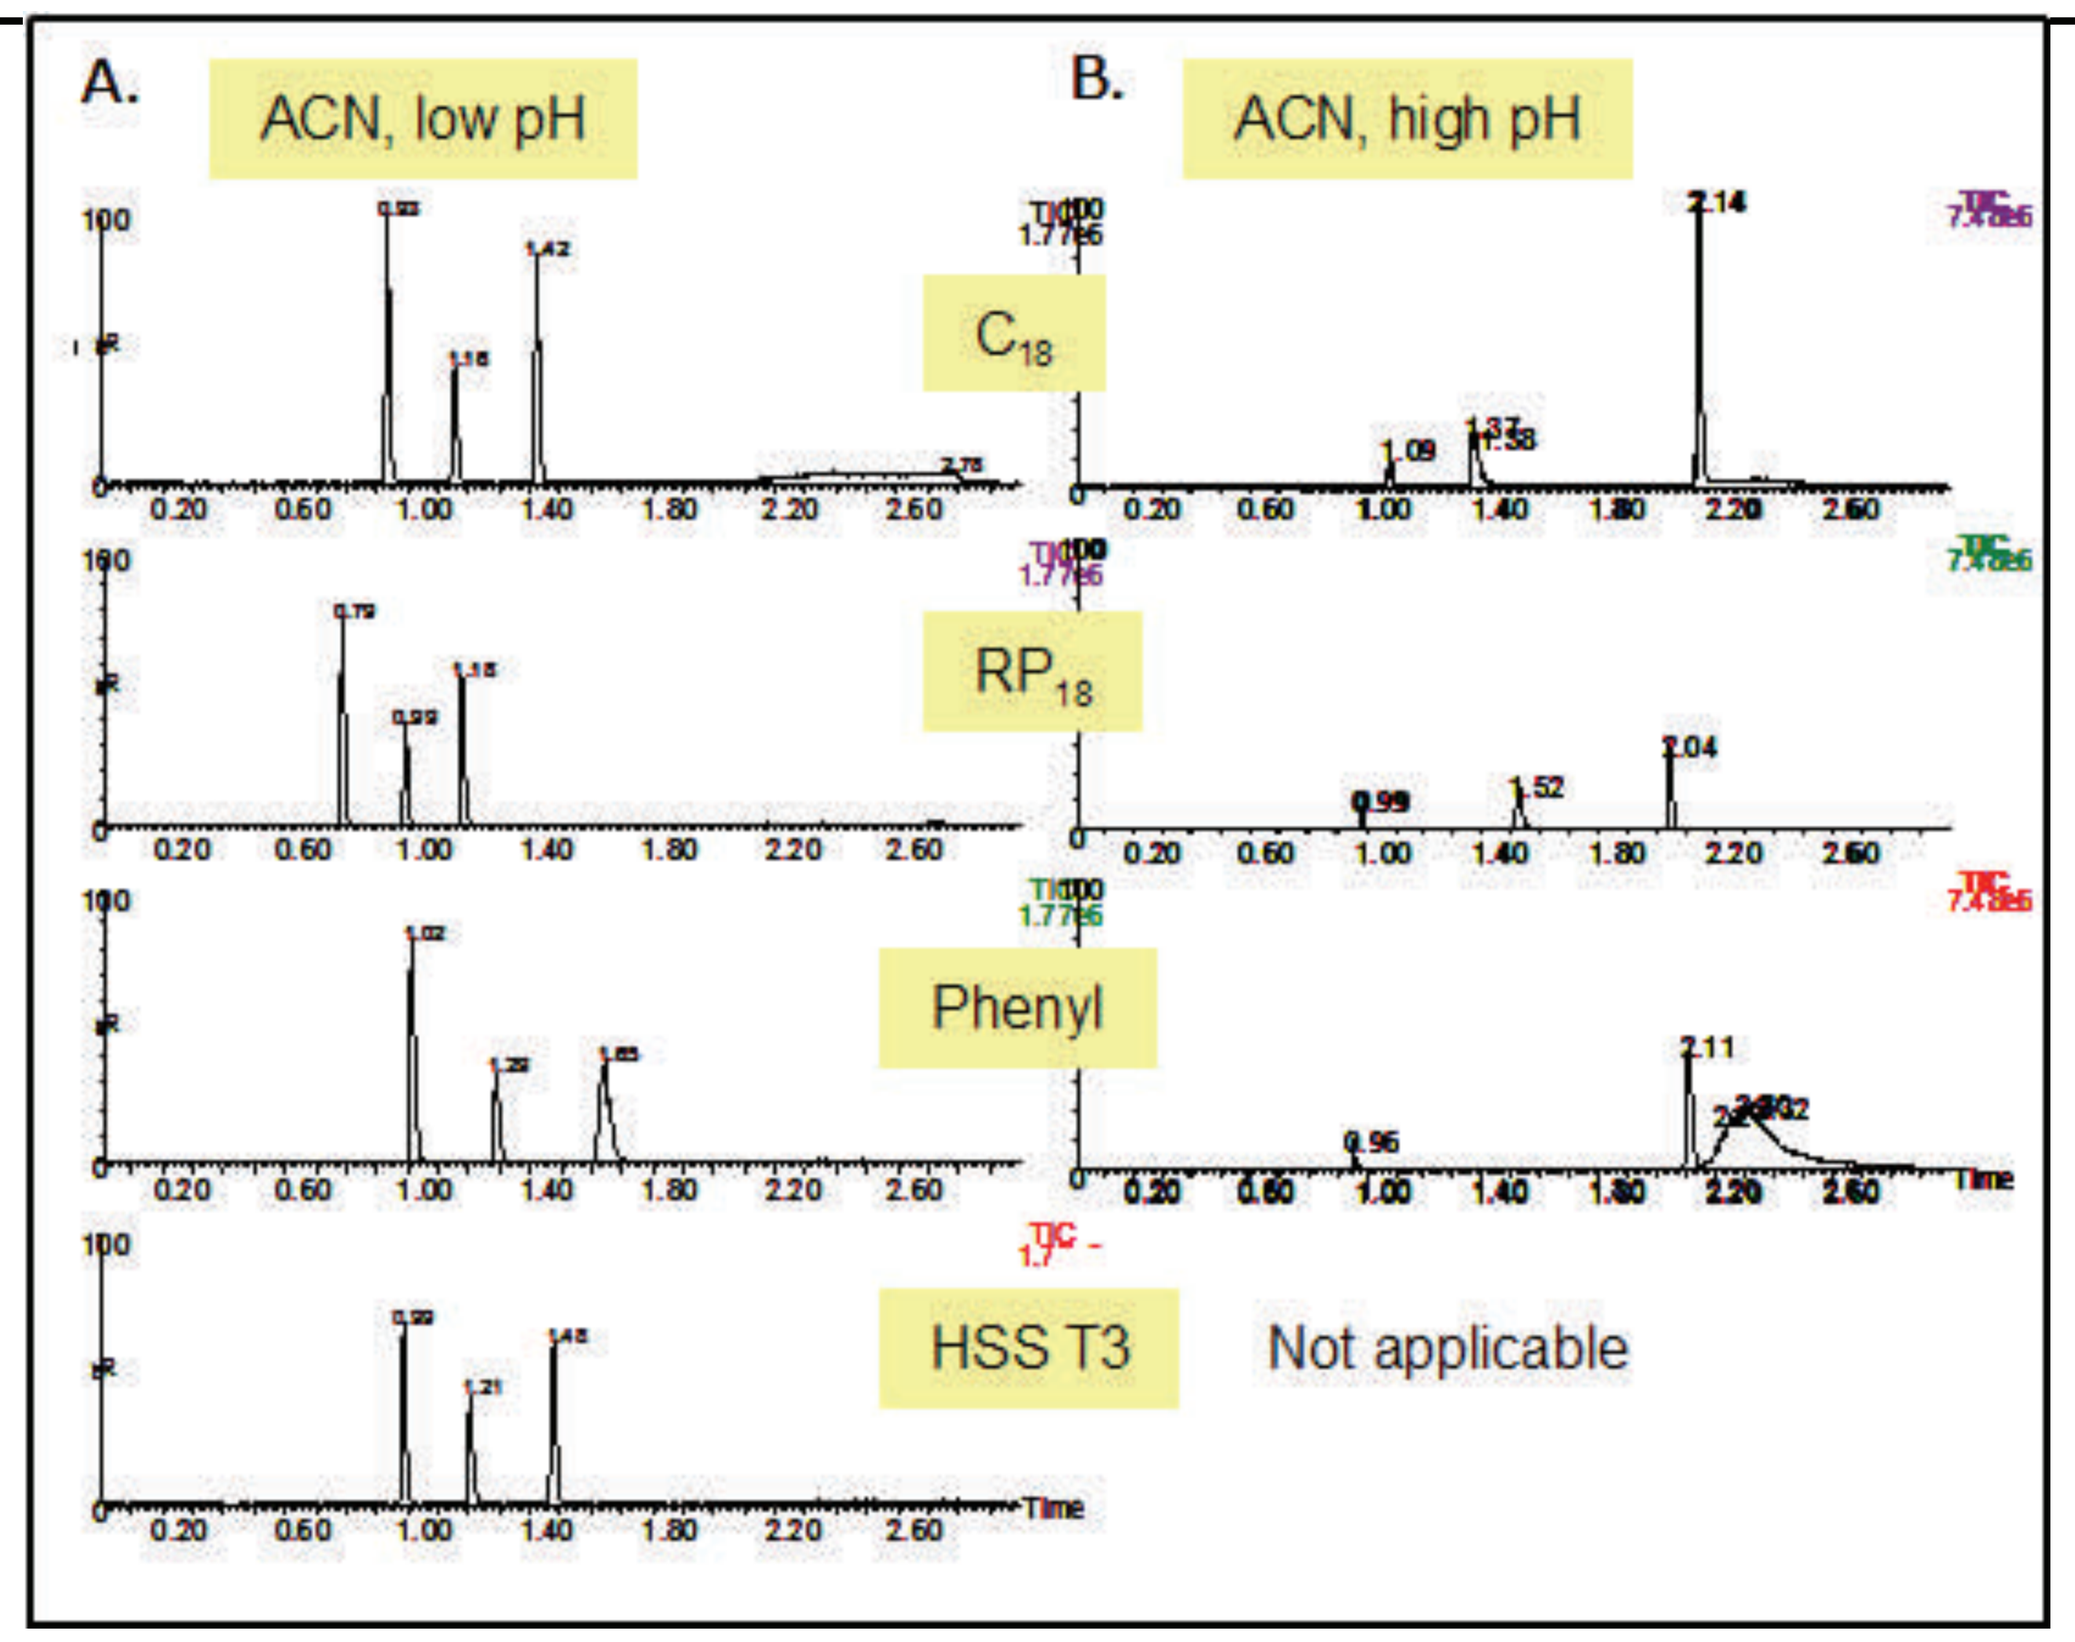

A.
ACN, low pH
B.
ACN, high pH
C18
RP18
Phenyl
HSS T3
Not applicable

Figure 3. MS chromatograms of 3 MRM transitions resulting from pH screening with ACN as the organic modifier.

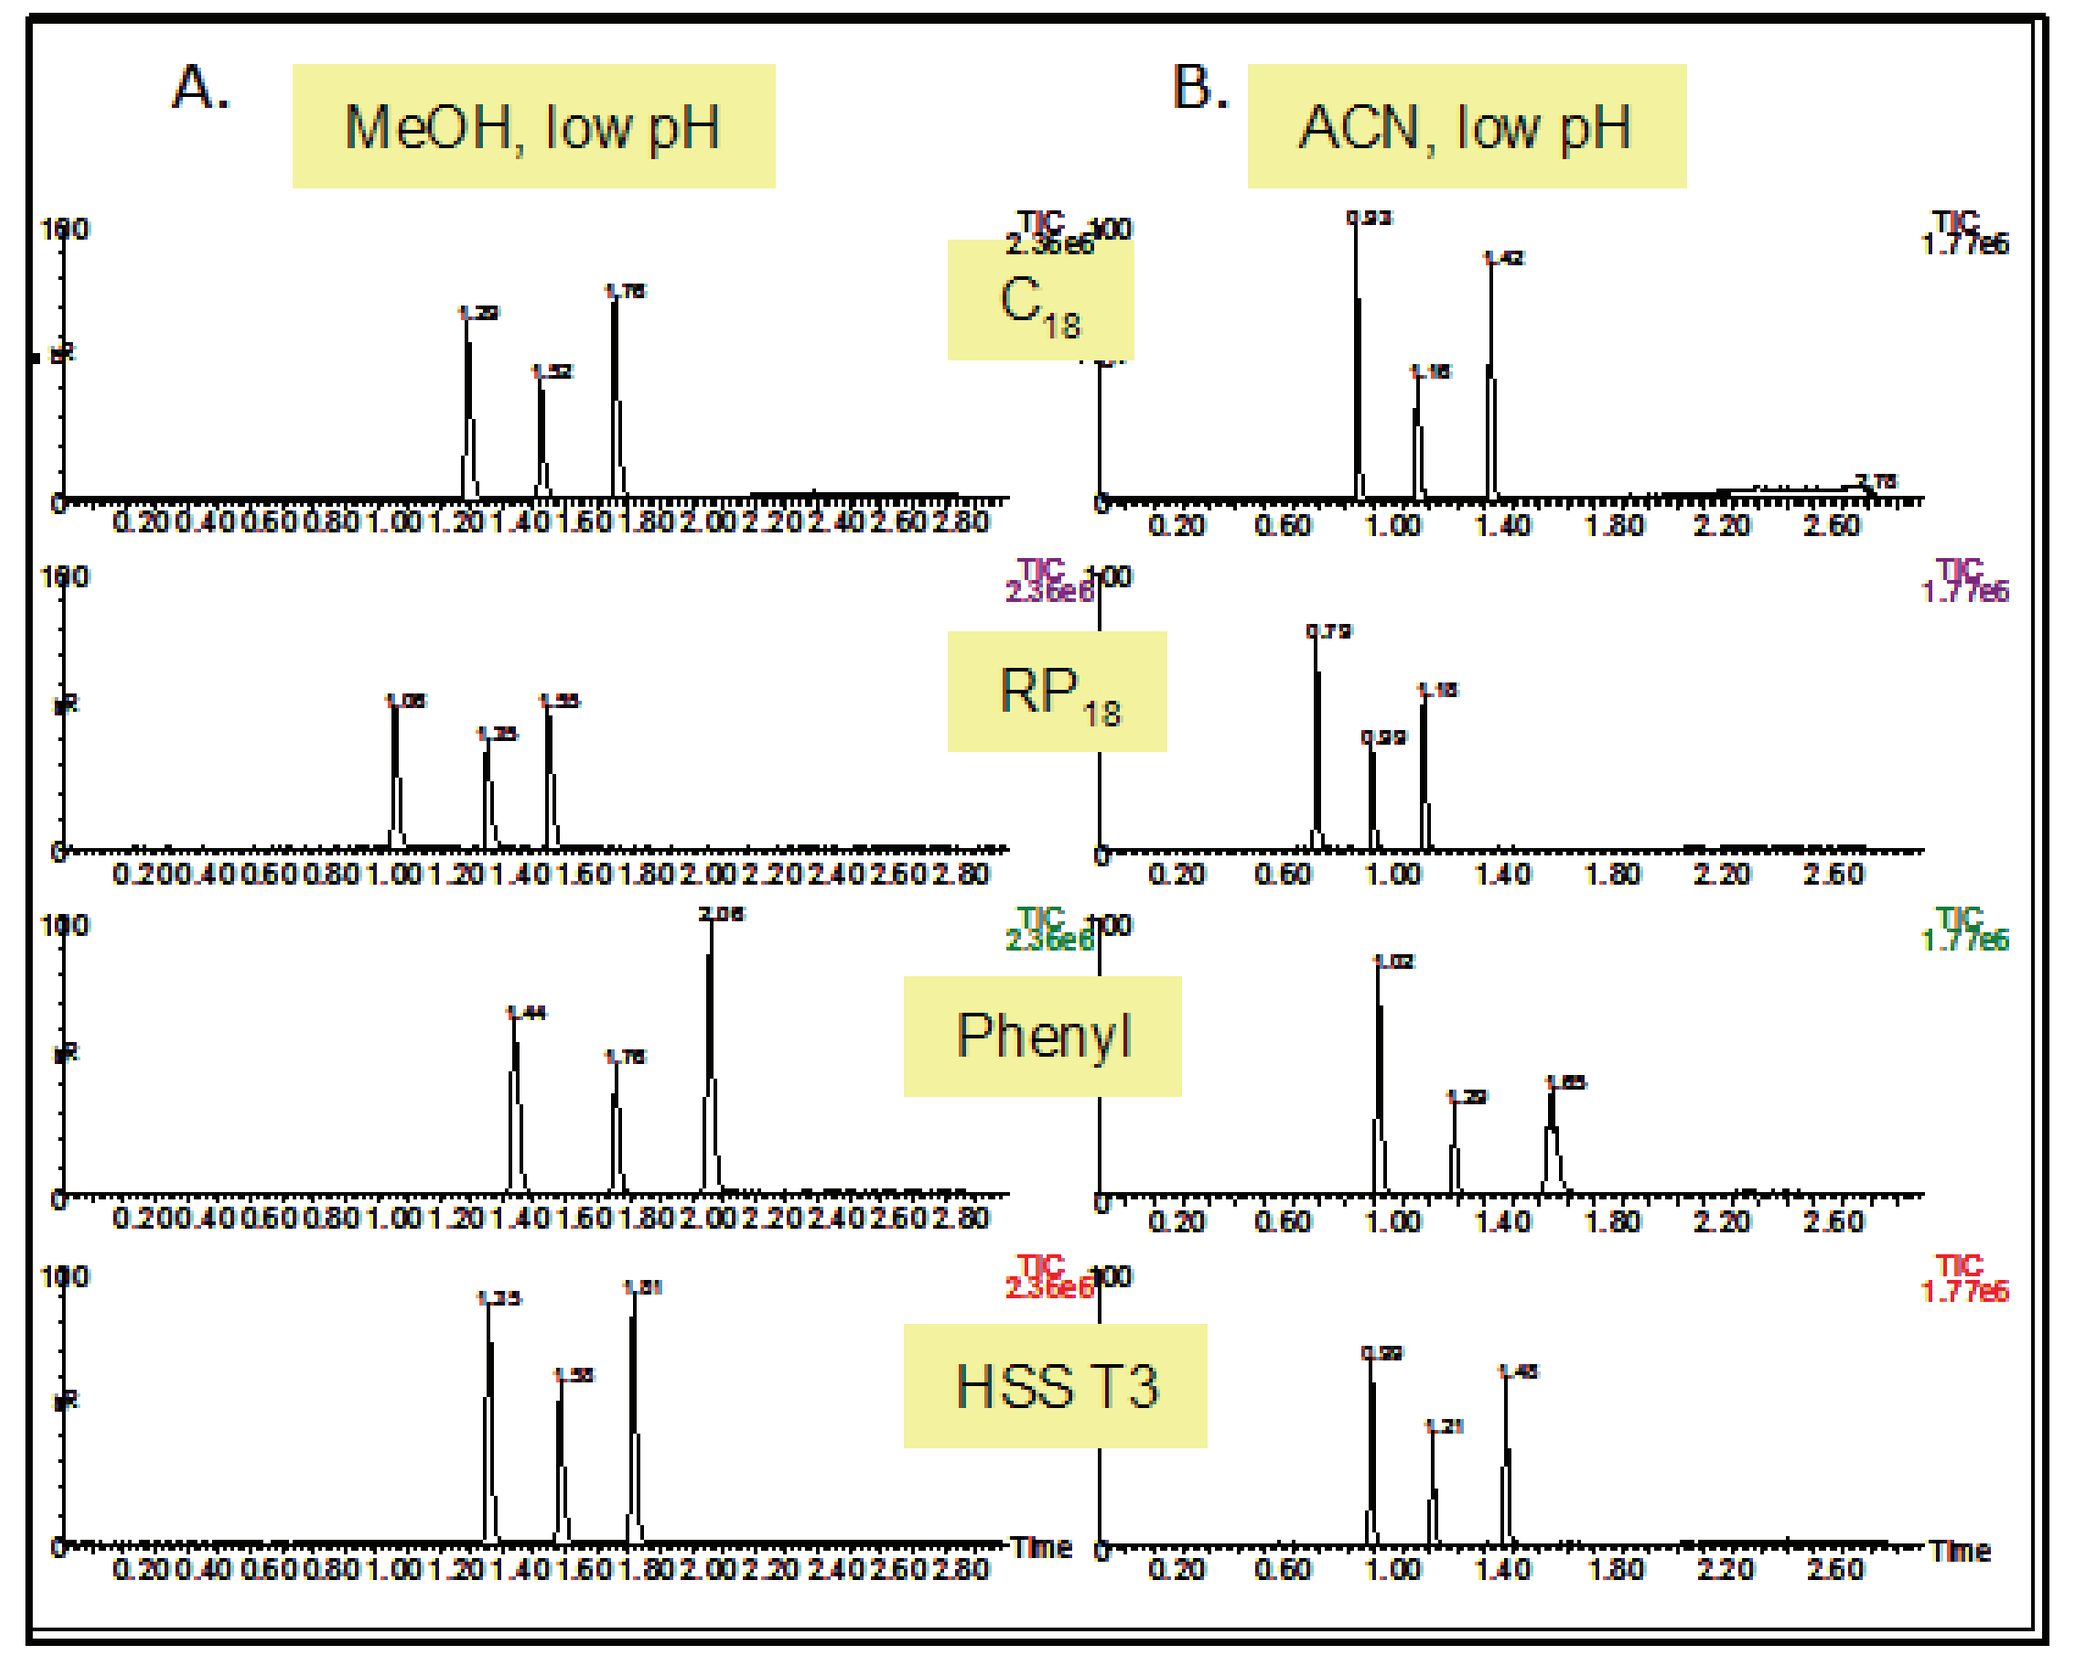

A.
MeOH, low pH
B.
ACN, low pH
C18
RP18
Phenyl
HSS T3

Figure 4. MS chromatograms of 3 MRM transitions resulting from organic modifier screening at low pH.

SPE SCREENING AND RECOVERY

Recovery of fexofenadine and terfenadine (we were unable to locate a new source of azacyclonol at screening time) from human urine were assessed by the comparison of peak areas of extracted samples containing 1 ng/mL fexofenadine and terfenadine to blank urine extracts spiked post-SPE at the same concentration. Recovery was calculated for both the Elute 1 fraction (100% organic, containing analyte bound by reversed-phase) and the Elute 2 fraction (containing analyte bound by ion-exchange) on all four of the mixed-mode sorbents. SPE recovery for terfenadine in urine is summarized in Figure 5. SPE recovery for fexofenadine in urine is summarized in Figure 6. Oasis® MCX, the mixed-mode strong cation exchange, was chosen as the starting point for further optimization, if necessary, as recoveries for both analytes were >85% in a single elution.

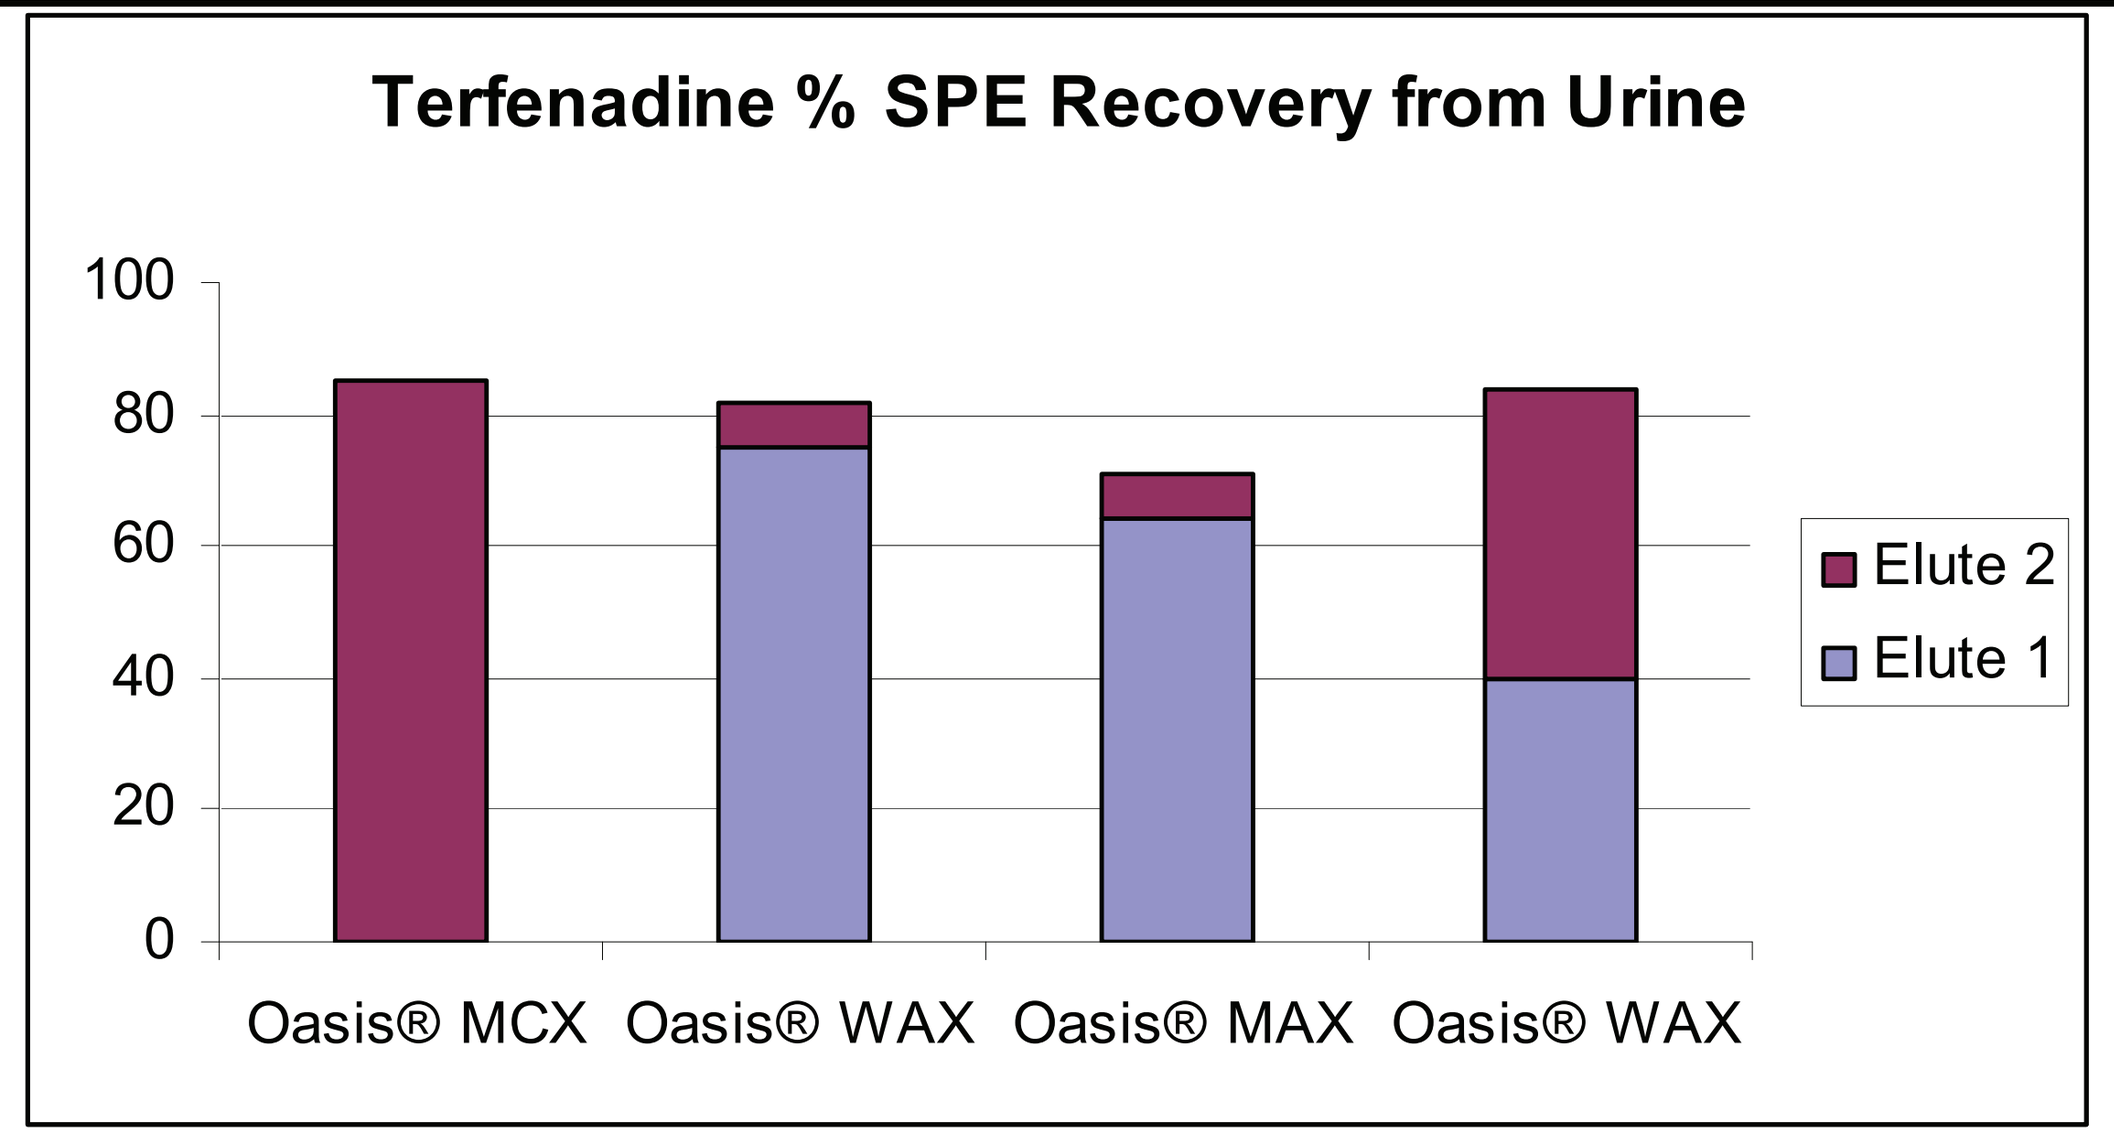

Terfenadine % SPE Recovery from Urine
100
80
60
40
20
0
Oasis® MCX
Oasis® WAX
Oasis® MAX
Oasis® WAX
Elute 2
Elute 1

Figure 5. % SPE recoveries for terfenadine extracted from human urine. Elute 1 = reversed-phase elution, Elute 2 = ion exchange elution

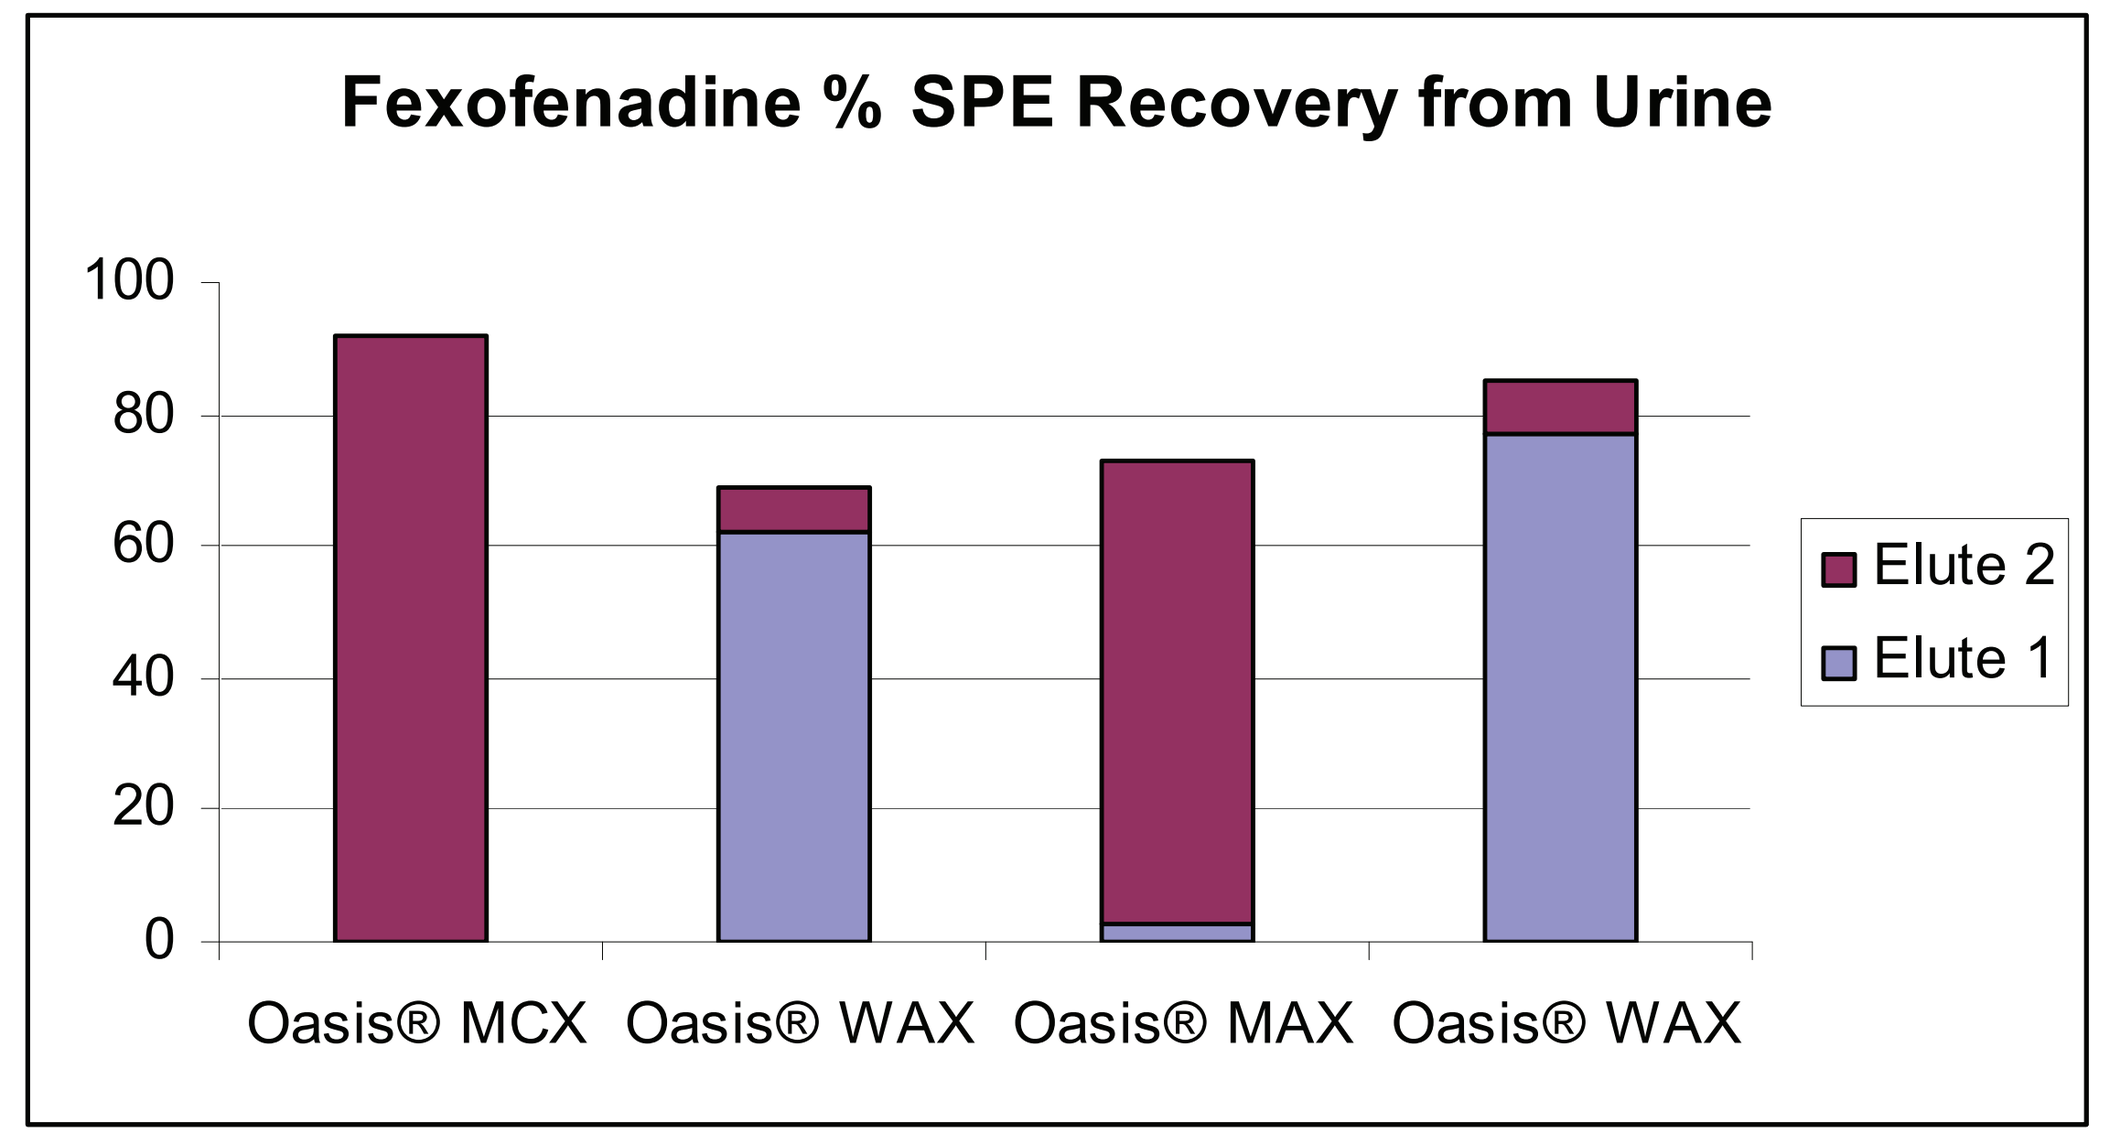

Fexofenadine % SPE Recovery from Urine
100
80
60
40
20
0
Oasis® MCX
Oasis® WAX
Oasis® MAX
Oasis® WAX
Elute 2
Elute 1

Figure 6. % SPE recoveries for fexofenadine extracted from human urine. Elute 1 = reversed-phase elution, Elute 2 = ion exchange elution

CONCLUSIONS

A straightforward approach to LC method development was successfully employed to determine optimum column chemistry, pH, and organic modifier for terfenadine and two of its metabolites. This method will be further optimized to shorten run time and will then become part of an overall bioanalytical method for these compounds. The entire screen consisting of 4 columns, 2 pHs, and 2 organic modifiers, took 49 minutes. Similarly, a straightforward SPE screening strategy was used to quickly identify the best starting SPE sorbent and protocol.

TO DOWNLOAD A COPY OF THIS POSTER, VISIT WWW.WATERS.COM/POSTERS